Helicobacter pylori Eradication Rates with Vonoprazan: Subgroup Analysis from the Phase 3 pHalcon-HP Trial

Sa1373

William D. Chey,[1] Francis Mégraud,[2] Loren Laine,[3,4] Neila Smith,[5] Eckhard Leifke,[5] Barbara Hunt,[5] Colin W. Howden[6]

[1]Department of Internal Medicine, Division of Gastroenterology and Hepatology, Michigan Medicine, Ann Arbor, MI, USA; [2]INSERM U1312, University of Bordeaux, Bordeaux, France; [3]Yale School of Medicine, New Haven, CT, USA; [4]VA Connecticut Healthcare System, West Haven, CT, USA; [5]Research and Development, Phathom Pharmaceuticals, Buffalo Grove, IL, USA; [6]Department of Medicine, University of Tennessee College of Medicine, Memphis, TN, USA

BACKGROUND

- Vonoprazan, a potassium-competitive acid blocker (P-CAB) is approved in the United States for the treatment of *Helicobacter pylori* infection in adults.[1]
- The rapid and potent acid suppression induced by vonoprazan enhances bacterial replication and the activity of some antibiotics used in the treatment of *H. pylori* infection.[2–5]
- pHalcon-HP was a large phase 3 trial that demonstrated superiority of vonoprazan dual and triple regimens over traditional lansoprazole-based triple therapy in treatment-naïve patients with *H. pylori* infection (NCT04167670).[6]
 - The vonoprazan triple therapy group had an *H. pylori* eradication rate of 80.8%, versus 68.5% for lansoprazole triple therapy (difference 12.3%; 95% CI, 5.7 to 18.8; superiority P<0.001).
 - The vonoprazan dual therapy group had an *H. pylori* eradication rate of 77.2% (difference to lansoprazole triple therapy 8.7%; 95% CI, 1.9 to 15.4; superiority P=0.013).
- Here, we present subgroup analyses from the pHalcon-HP trial assessing differences in *H. pylori* eradication rates among treatment groups related to baseline demographic and clinical characteristics of patients in the study.

METHODS

- pHalcon-HP enrolled treatment-naïve patients (≥18 years old) with *H. pylori* infection, confirmed by a positive ^{13}C-urea breath test (UBT) (BreathTek; Otsuka America Pharmaceutical, Inc, Rockville, MD) during the screening period, and with ≥1 of the following clinical conditions:
 - Dyspepsia, a recent/new diagnosis of non-bleeding peptic ulcer, history of peptic ulcer not previously treated for *H. pylori*, or requirement for long-term, non-steroidal anti-inflammatory drug treatment at a stable dose.
- During the screening period, patients underwent endoscopy and two gastric mucosal biopsies (greater curve of the antrum and lesser curve of the body) were taken for culture and antimicrobial susceptibility testing.
- Patients were randomized 1:1:1 to 14 days of treatment with either (**Figure 1**):
 - Open-label vonoprazan dual therapy with vonoprazan 20 mg twice daily and amoxicillin 1 g 3 times daily, or
 - Double-blind vonoprazan triple therapy with vonoprazan 20 mg, amoxicillin 1 g, and clarithromycin 500 mg, twice daily, or
 - Double-blind lansoprazole triple therapy with lansoprazole 30 mg, amoxicillin 1 g, and clarithromycin 500 mg, twice daily
- *H. pylori* status was assessed by ^{13}C-UBT 4 weeks after the last dose of study drug.
- Additional analyses assessed *H. pylori* eradication rates in subgroups based on baseline demographic and clinical characteristics.

Figure 1. pHalcon-HP study design

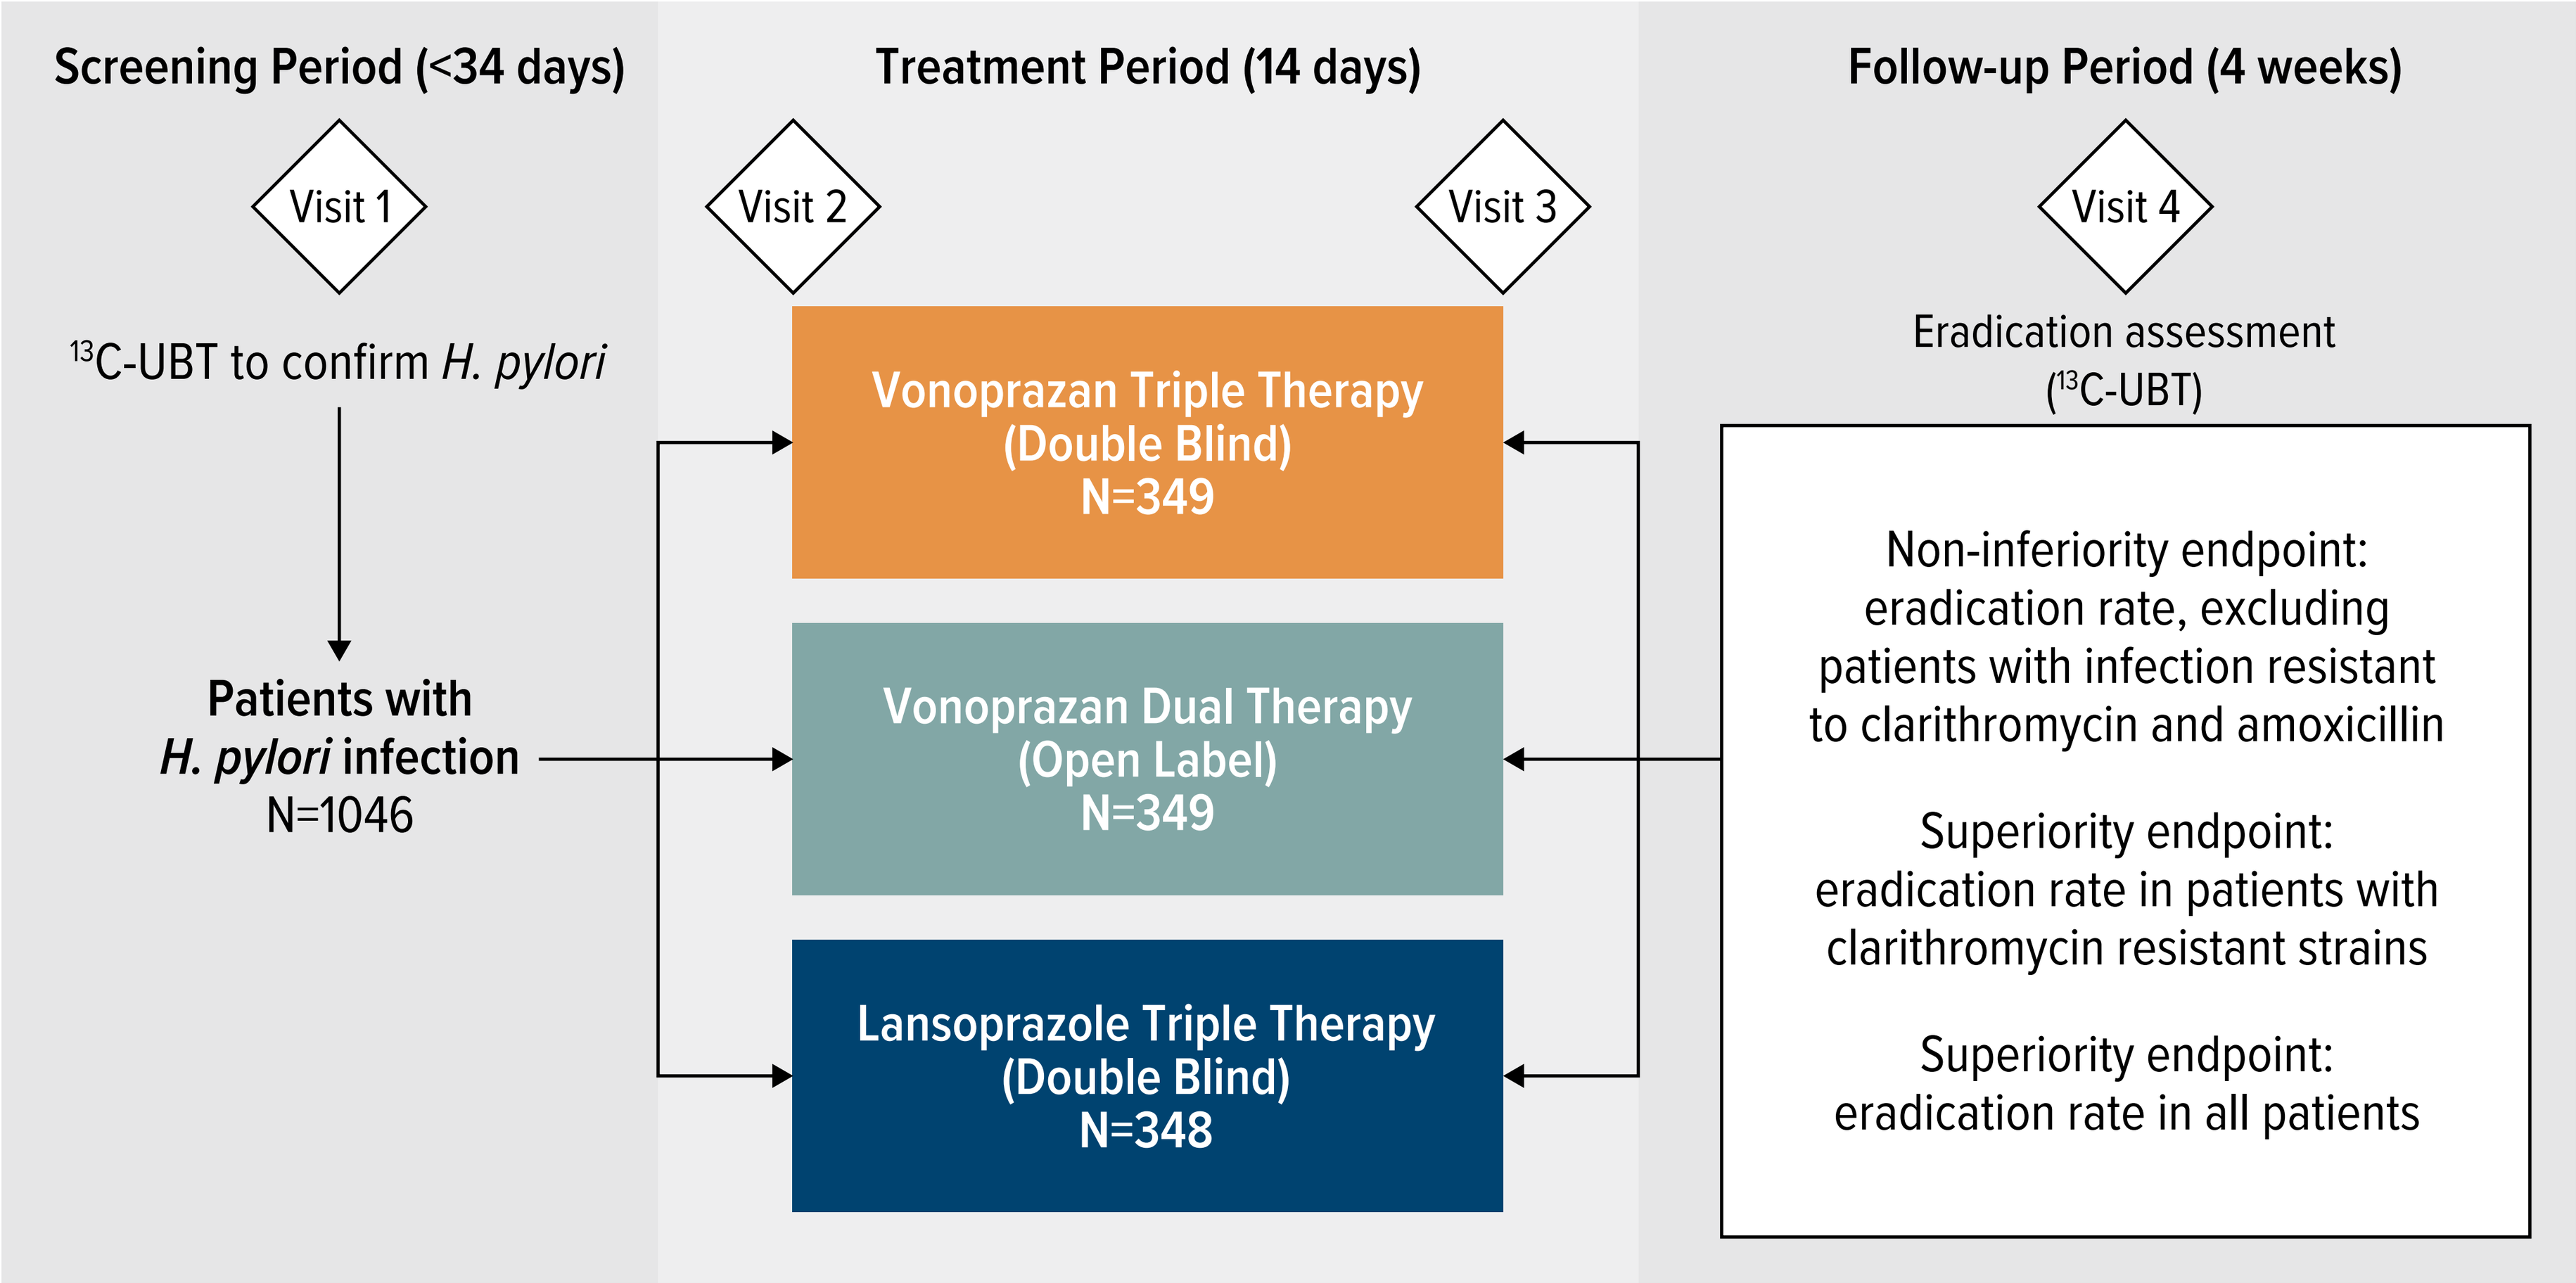
Screening Period (<34 days)
Treatment Period (14 days)
Follow-up Period (4 weeks)
Visit 1
Visit 2
Visit 3
Visit 4
13C-UBT to confirm H. pylori
Patients with H. pylori infection N=1046
Vonoprazan Triple Therapy (Double Blind) N=349
Vonoprazan Dual Therapy (Open Label) N=349
Lansoprazole Triple Therapy (Double Blind) N=348
Eradication assessment (13C-UBT)
Non-inferiority endpoint: eradication rate, excluding patients with infection resistant to clarithromycin and amoxicillin
Superiority endpoint: eradication rate in patients with clarithromycin resistant strains
Superiority endpoint: eradication rate in all patients

^{13}C-UBT, ^{13}C-urea breath test.

RESULTS

Baseline characteristics

- 992 patients were included in this analysis.
- Baseline characteristics were similar among the three treatment groups (**Table**).

Table. Baseline clinical and demographic characteristics of patients enrolled in the pHalcon-HP study

Characteristic	Vonoprazan dual therapy (n=324)	Vonoprazan triple therapy (n=338)	Lansoprazole triple therapy (n=330)
Age, years (SD)	51.8 (13.6)	50.6 (13.9)	51.8 (13.5)
Sex, n (%)			
Male	128 (39.5)	118 (34.9)	125 (37.9)
Female	196 (60.5)	220 (65.1)	205 (62.1)
Race, n (%)			
American Indian or Alaska Native	0	1 (0.3)	1 (0.3)
Asian	3 (0.9)	6 (1.8)	6 (1.8)
Black or African American	21 (6.5)	29 (8.6)	22 (6.7)
Native Hawaiian or Other Pacific Islander	1 (0.3)	0	0
White	294 (90.7)	298 (88.2)	297 (90.0)
Other	3 (0.9)	1 (0.3)	3 (0.9)
Ethnicity, n (%)			
Hispanic or Latino	86 (26.5)	94 (27.8)	80 (24.2)
BMI, median (Q1, Q3)	28.7 (25.0, 32.8)	29.1 (25.7, 32.9)	28.9 (25.7, 32.2)
Region, n (%)			
United States	134 (41.4)	144 (42.6)	134 (40.6)
Europe	190 (58.6)	194 (57.4)	196 (59.4)

BMI, body mass index.

Subgroup analysis

- The magnitude of the treatment effects for vonoprazan dual and triple therapy versus lansoprazole triple therapy were generally similar across subgroups based on baseline demographic and clinical characteristics (**Figures 2 and 3**).

Figure 2. Differences in *H. pylori* eradication rates between vonoprazan dual therapy and lansoprazole triple therapy, according to baseline demographics and clinical characteristics

Subgroup	Difference (%)	95% CI
Age (years)		
<45	20.5	7.60, 32.79
≥45 to <65	1.5	-8.36, 11.29
≥65	6.4	-6.98, 20.25
Sex		
Male	4.6	-6.48, 15.67
Female	11.3	2.63, 19.79
Race		
White	8.9	1.68, 15.98
Other†	6.3	-17.07, 28.46
Ethnicity		
Hispanic or Latino	8.4	-5.93, 22.59
Not Hispanic or Latino	9.2	1.45, 16.80
Region		
United States	6.7	-4.23, 17.55
Europe	10.1	1.37, 18.69
Baseline BMI category (kg/m²)		
<25	10.5	-2.80, 24.37
≥25 to <30	8.2	-3.76, 19.82
>30	6.8	-3.73, 17.31

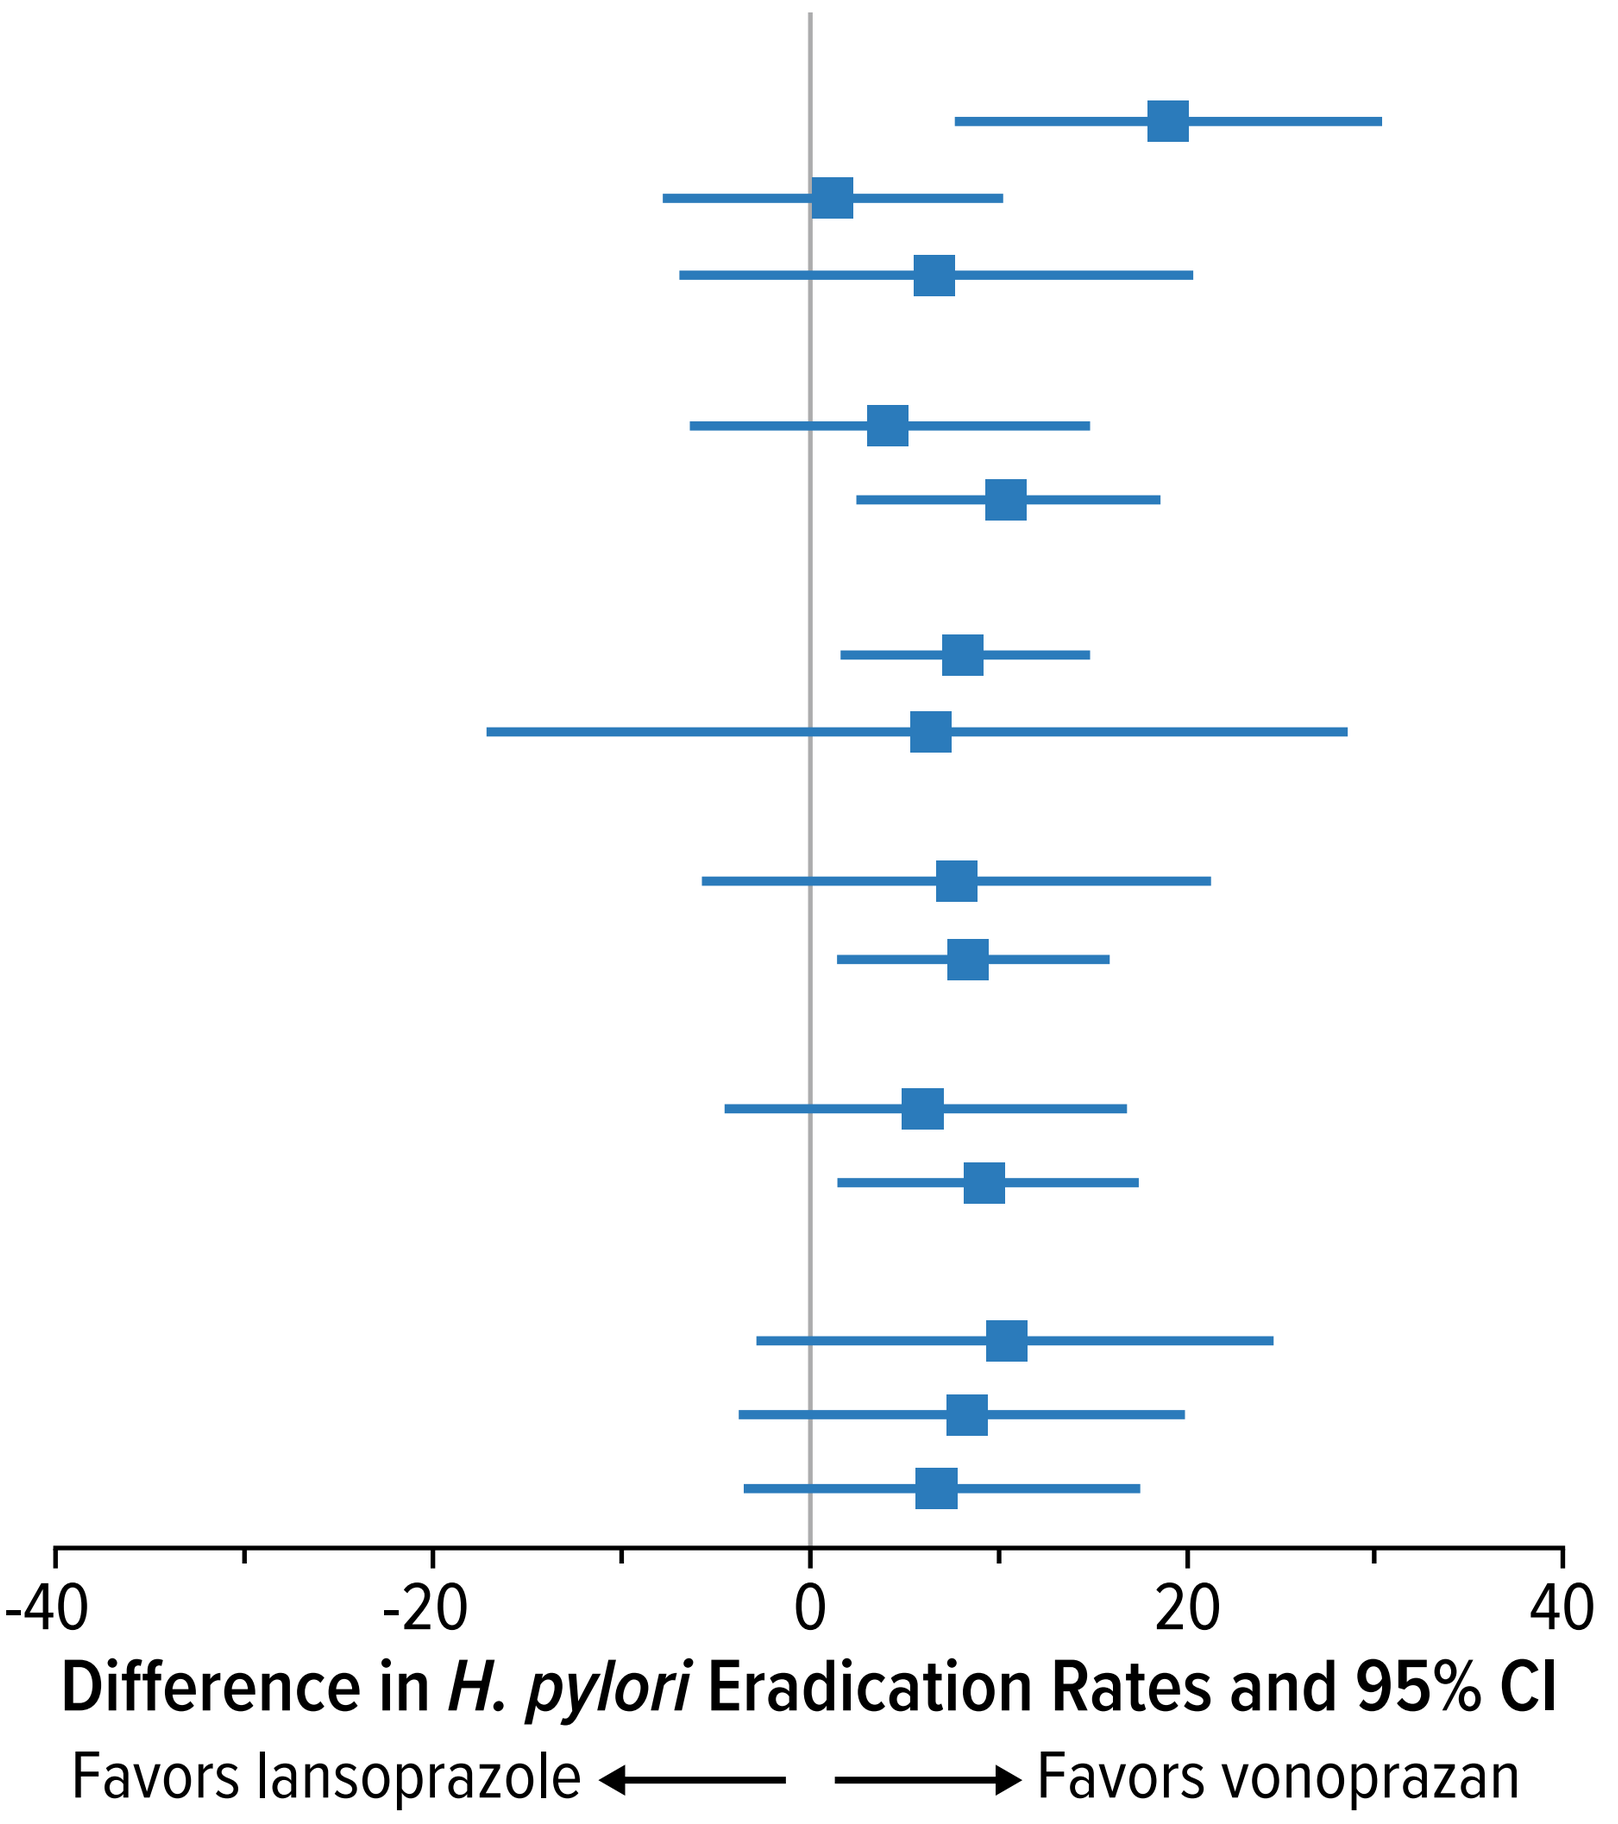
-40
-20
0
20
40
Difference in H. pylori Eradication Rates and 95% CI
Favors lansoprazole
Favors vonoprazan

†Other race includes: Black or African American, Asian, American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander.
BMI, body mass index; CI, confidence interval; NA, not applicable.

Figure 3. Differences in *H. pylori* eradication rates between vonoprazan triple therapy and lansoprazole triple therapy, according to baseline demographics and clinical characteristics

Subgroup	Difference (%)	95% CI
Age (years)		
<45	25.5	13.80, 36.91
≥45 to <65	1.0	-9.23, 10.98
≥65	14.4	2.37, 27.33
Sex		
Male	10.9	-0.02, 21.62
Female	13.1	4.83, 21.34
Race		
White	12.9	5.91, 19.74
Other†	9.6	-11.28, 30.55
Ethnicity		
Hispanic or Latino	8.8	-5.22, 22.70
Not Hispanic or Latino	14.4	7.08, 21.61
Region		
United States	7.1	-3.57, 17.81
Europe	16.2	7.99, 24.31
Baseline BMI category (kg/m²)		
<25	7.9	-6.04, 22.14
≥25 to <30	16.8	5.64, 27.52
>30	10.4	0.35, 20.45

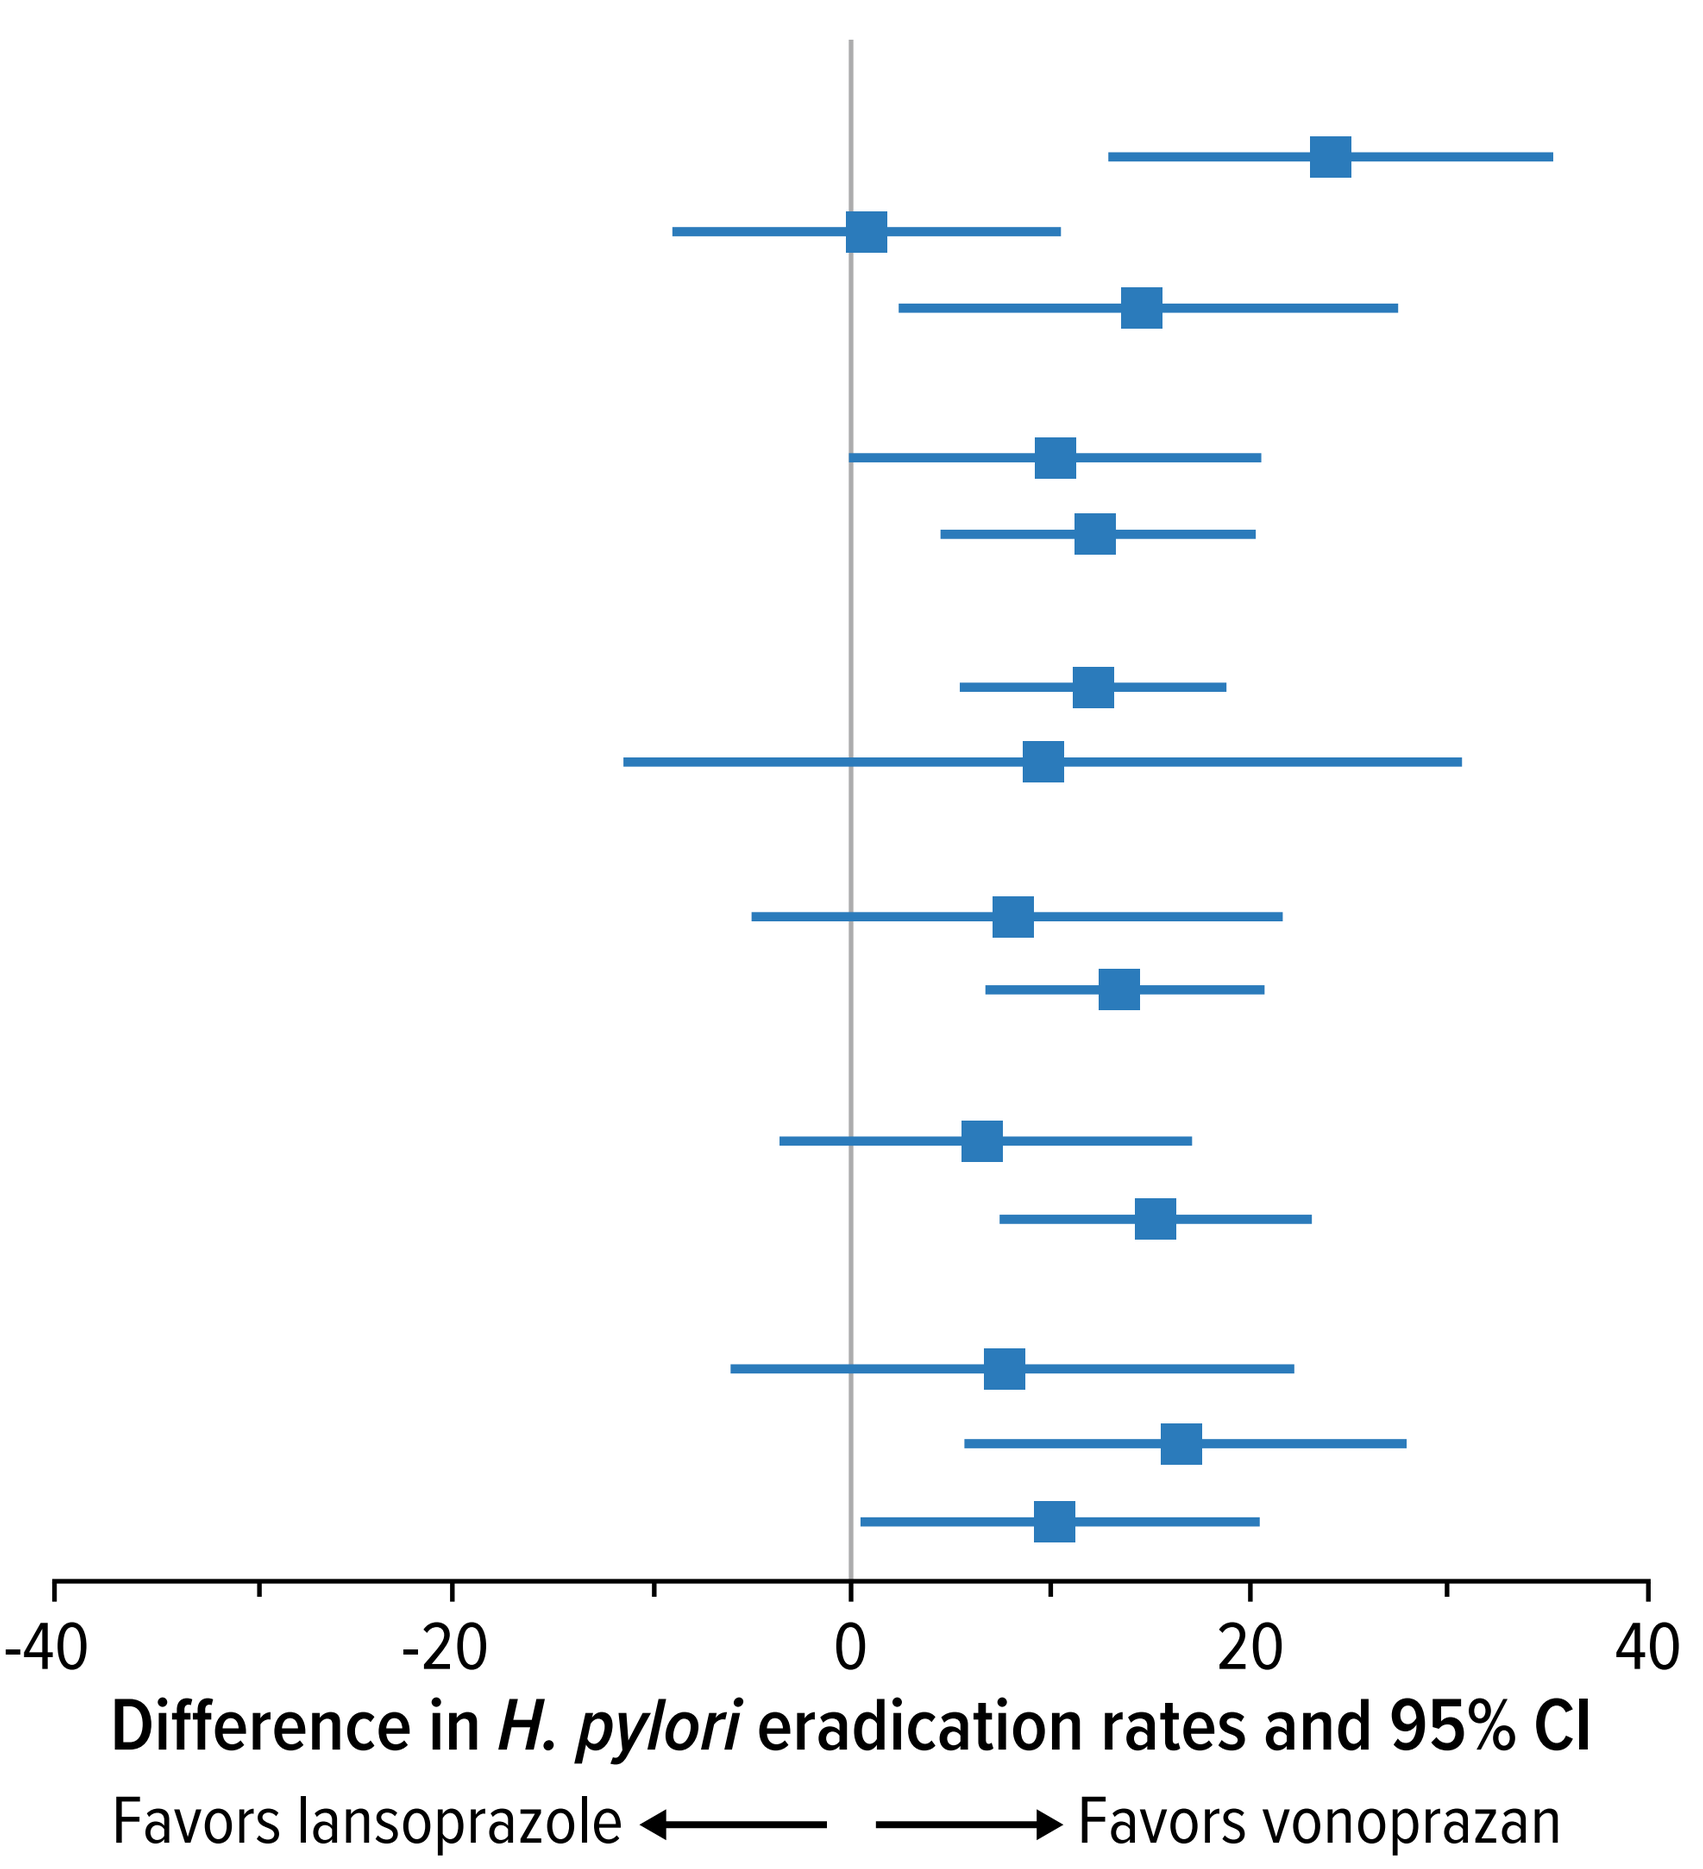
-40
-20
0
20
40
Difference in H. pylori eradication rates and 95% CI
Favors lansoprazole
Favors vonoprazan

†Other race includes: Black or African American, Asian, American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander.
BMI, body mass index; CI, confidence interval; NA, not applicable

CONCLUSIONS

- **The higher *H. pylori* eradication rates with vonoprazan-based regimens versus lansoprazole triple therapy were generally similar regardless of different baseline characteristics including age, geographic region, race, ethnicity, sex and BMI.**

Acknowledgments

Medical writing support was provided by Samantha Agron, MD of Synergy Medical Communications, funded by Phathom Pharmaceuticals in accordance with Good Publications Practice (GPP 2022) guidelines.

Conflicts of interest

WDC has served as a consultant for AbbVie, BioAmerica, Ironwood Pharmaceuticals, QOL Medical, Nestle, Phathom Pharmaceuticals, RedHill Biopharma, Salix/Valeant, Takeda, Urovant, and Vibrant; has received grant/research support from Commonwealth Diagnostics International, QOL Medical, Salix; has stock/stock options from Dieta, Kiwi Biosciences, Isothrive, and Modify Health; and patents relating to methods and kits for identifying food sensitivities and intolerances, digital manometry, and a rectal expulsion device. **FM** has served on an advisory committee member for Phathom Pharmaceuticals. **LL** has served as a consultant for Phathom Pharmaceuticals. **NS, EL,** and **BH** are employees of Phathom Pharmaceuticals. **EL** also discloses stockholder interest in Phathom Pharmaceuticals. **CWH** has served as a consultant for Phathom Pharmaceuticals, RedHill Biopharma, Ironwood Pharmaceuticals and Allakos, and as a speaker for RedHill Biopharma, Alnylam Pharmaceuticals and Sanofi/Genzyme; he also owns stock in Antibe Therapeutics.

References

1. Vonoprazan [package insert]. Buffalo Grove, IL: Phathom Pharmaceuticals, Inc.; 2022.
2. Erah PO et al. *J Antimicrob Chemother* 1997;39:5–12.
3. Ke H et al. *Helicobacter* 2021;26:e12768.
4. Takeuchi T et al. *Aliment Pharmacol Ther* 2020;51:534–43.
5. Laine L et al. *Am J Gastroenterol* 2022;117:1158–61.
6. Chey WD et al. *Gastroenterology* 2022;163:609–19.

Funding details

This study was sponsored by Phathom Pharmaceuticals.