A Model-Based Meta-Analysis of the Relationship Between pH Control and Erosive Esophagitis Healing Rates

Tu1296

Colin W. Howden,[1] Carmelo Scarpignato,[2] Eckhard Leifke,[3] Darcy Mulford,[3] Gezim Lahu,[4] Axel Facius,[4] Yuhong Yuan,[5,6] Richard Hunt[5,6]

[1]University of Tennessee College of Medicine, Memphis, TN, USA; [2]Department of Health Sciences, United Campus of Malta, Msida, MSD 9024, Malta; [3]Phathom Pharmaceuticals, Research and Development, Buffalo Grove, IL, USA; [4]thinkQ[2] AG, Baar, Switzerland; [5]Department of Medicine, Division of Gastroenterology, McMaster University, Hamilton, ON, Canada; [6]Farncombe Family Digestive Health Research Institute, McMaster University, Hamilton, ON, Canada

BACKGROUND

- Optimized acid suppression is critical for healing of erosive esophagitis.[1–3]
- However, acid-suppressing drugs vary in their pharmacodynamics (PD) and, consequently, their effectiveness.
- Maintenance of intragastric pH >4 for prolonged periods over the 24-hour day, known as pH >4 holding-time ratio (HTR), is associated with higher erosive esophagitis healing rates.[4,5]
- Current guidelines for erosive esophagitis predominantly recommend proton pump inhibitors (PPIs), with only a limited role for H_2-receptor antagonists (H_2RAs). However, some patients experience incomplete healing.[6–8]
- Potassium-competitive acid blockers (P-CABs), which are associated with significantly higher pH HTRs than PPIs, may provide better rates of erosive esophagitis healing.[9–14]
- To understand the significance of differing PD profiles, gastric antisecretory activity must be placed in the context of clinical improvement.
- The aim of this meta-analysis was to explore and model the relationship between pH HTRs and erosive esophagitis healing rates with H_2RAs, PPIs and P-CABs.

METHODS

Systematic literature search

- We conducted a comprehensive computer-aided literature search using MEDLINE and Embase (via Ovid) up to March 04, 2022 for fully published, English language studies in adults ≥18 years of age.
- Two data sets were established:
 1. Mean percentage of holding time (pH >4) from PD studies in healthy human participants investigating the gastric acid-suppressing effect of H_2RAs, PPIs or P-CABs at therapeutic doses and steady state (defined as between days 5 and 8 of daily dosing for the PPIs and P-CABs)
 2. Healing rates from randomized clinical trials (RCTs) investigating H_2RAs, PPIs or P-CABs in erosive esophagitis

Data analysis

- Individual study pH results at days 5–8 were pooled by drug dose and regimen using weighted means (weighted by size of the per-protocol population).
- Individual study healing rates were also pooled by week of assessment (week 4 and/or 8) using a generalized linear mixed model, specifically a random intercept regression model with a logit transformation.
- The two aggregated datasets for mean pH >4 holding times and erosive esophagitis healing rates were merged by drug and dose in order to explore the relationship between pH control in healthy participants with the healing rates obtained from the RCTs.
- The merged dataset further contained information on drug class (H_2RA, PPI or P-CAB) and approval status in the United States (US) and European Union (EU).
- A non-linear mixed-effects model was developed to characterize and quantify the relationship between pH >4 holding time (independent variable) and erosive esophagitis healing rates at weeks 4 and 8 (dependent variable).
- Both approved and non-approved treatments were included in the model development, but only approved treatments were included in the model simulations.

RESULTS

Systematic literature review

- The literature search identified 1,747 English language publications.
 - After screening, 181 relevant studies were identified (**Figure 1**), including 80 for pH HTR data and 101 for erosive esophagitis healing rates.

Figure 1. PRISMA diagram of study selection and disposition

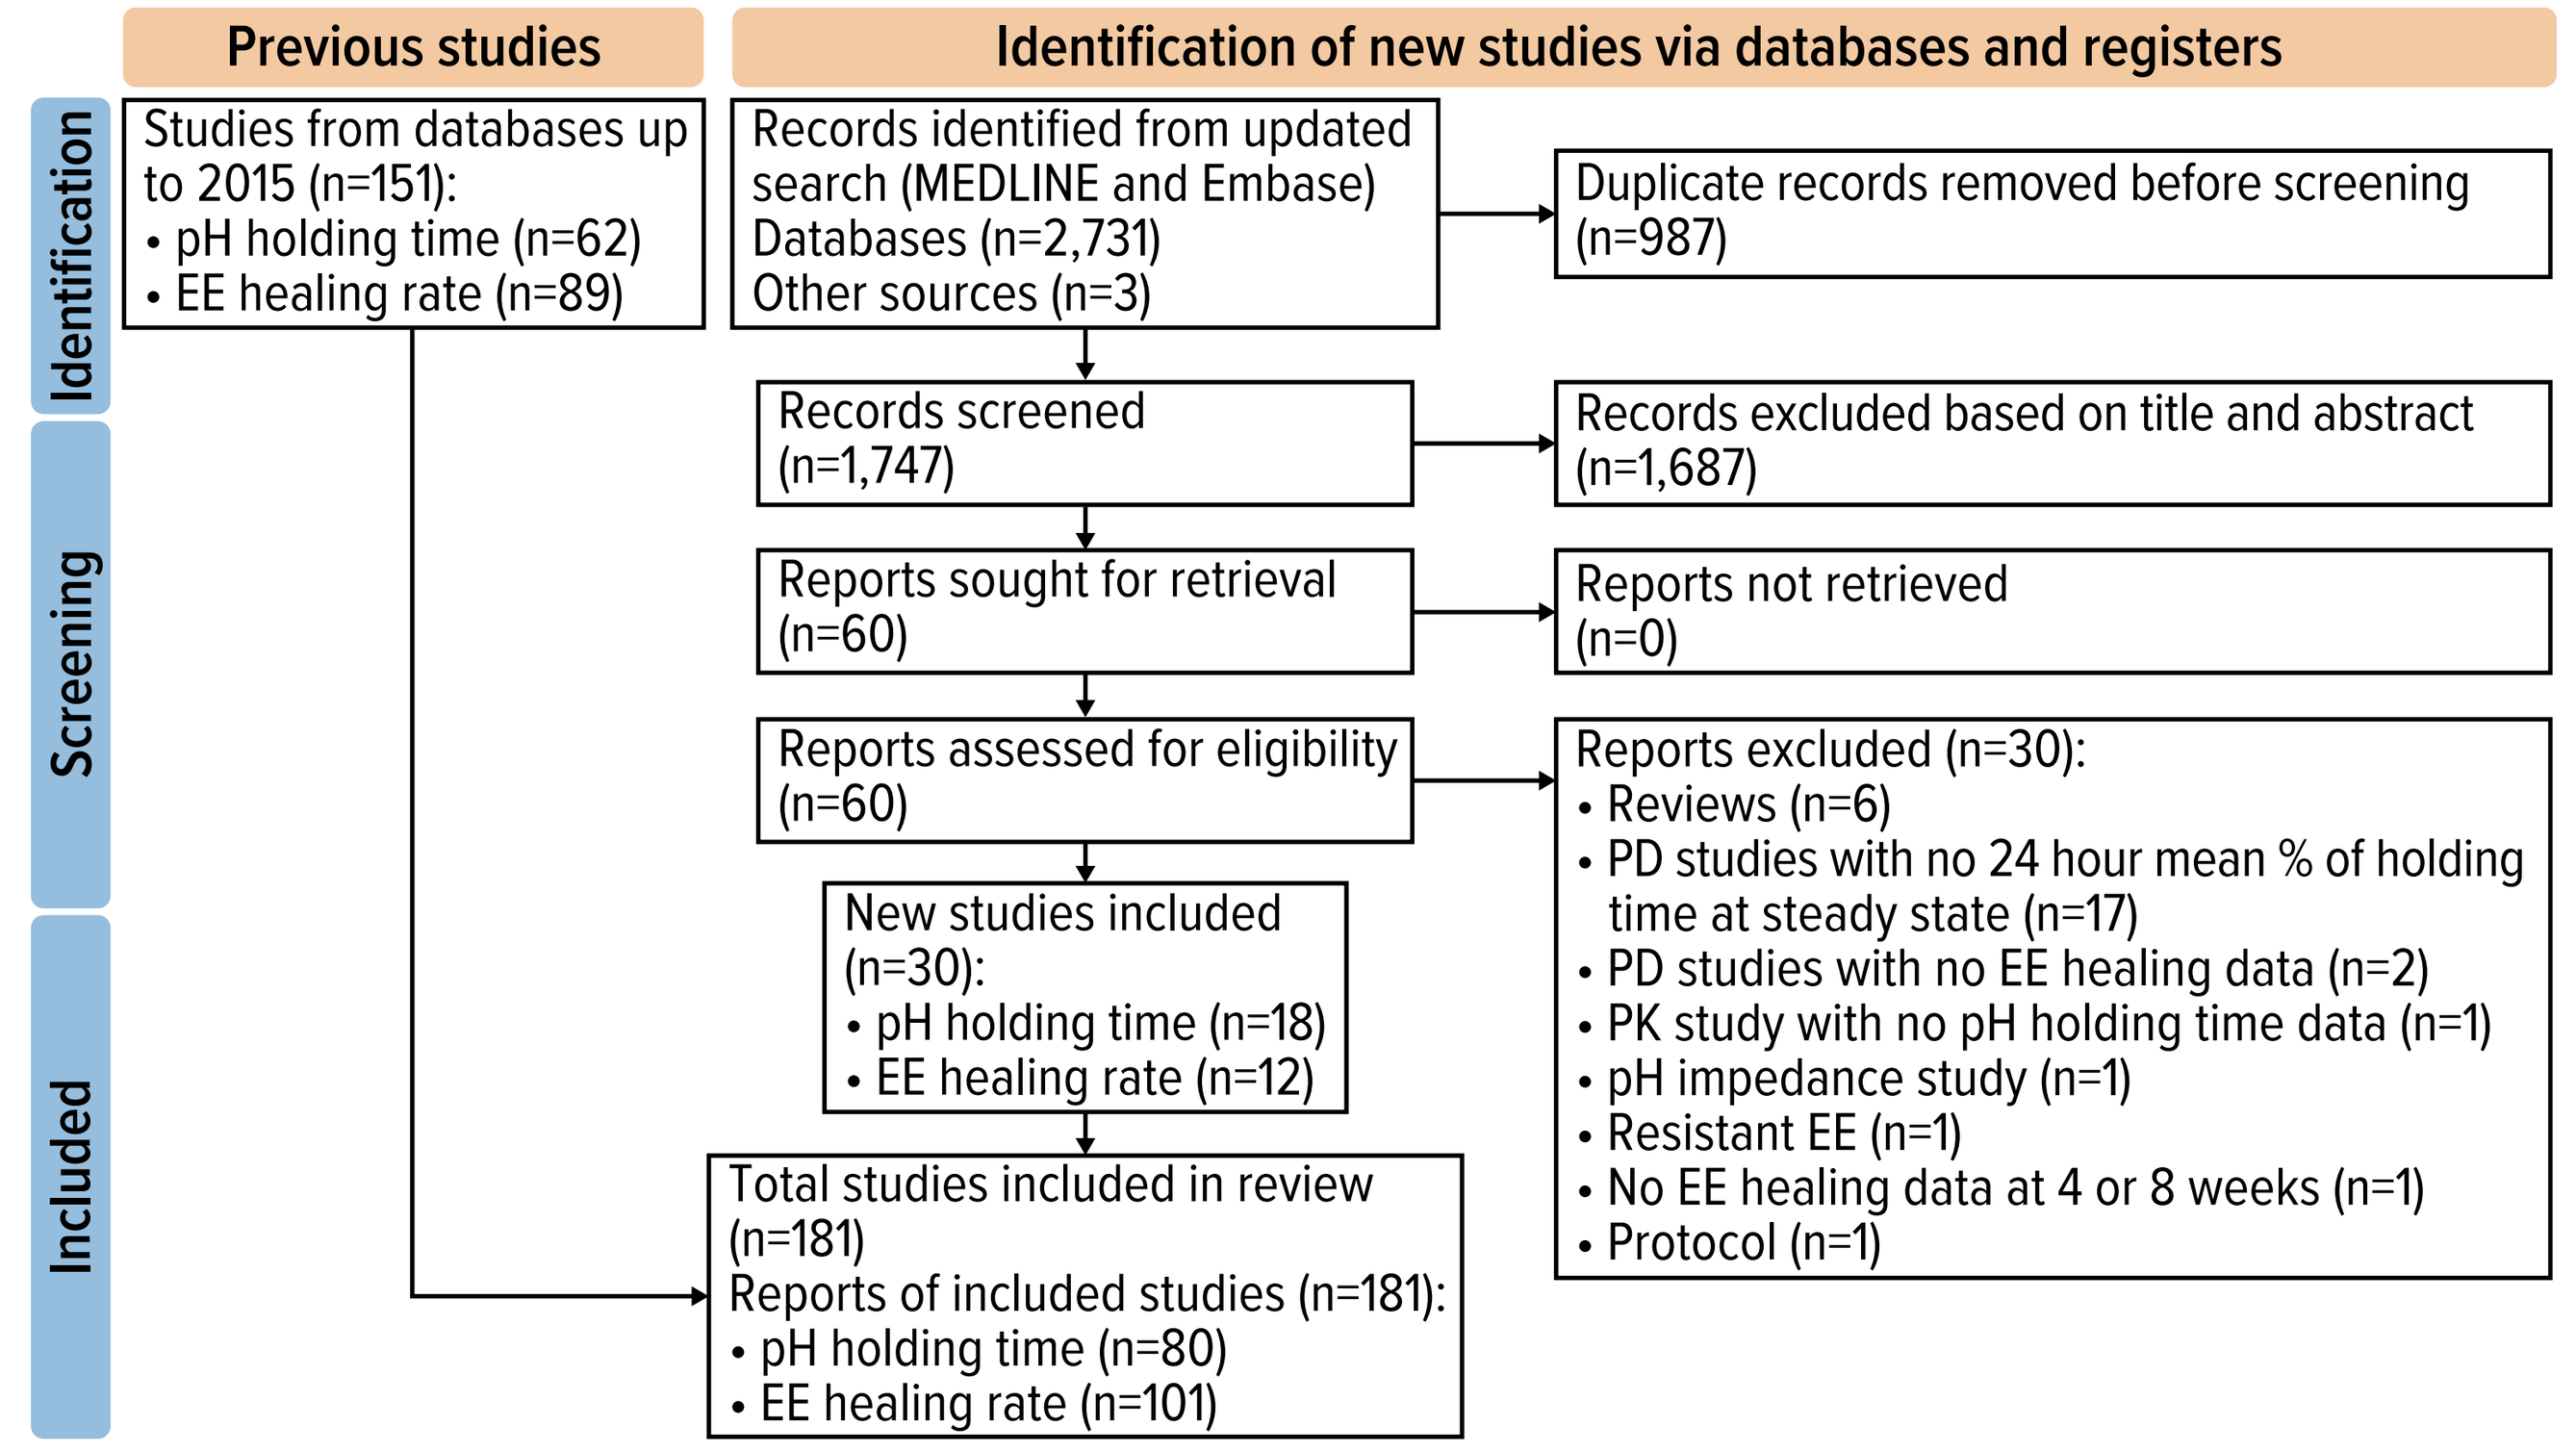
Previous studies
Identification of new studies via databases and registers
Identification
Screening
Included
Studies from databases up to 2015 (n=151):
• pH holding time (n=62)
• EE healing rate (n=89)
Records identified from updated search (MEDLINE and Embase)
Databases (n=2,731)
Other sources (n=3)
Duplicate records removed before screening (n=987)
Records screened (n=1,747)
Records excluded based on title and abstract (n=1,687)
Reports sought for retrieval (n=60)
Reports not retrieved (n=0)
Reports assessed for eligibility (n=60)
Reports excluded (n=30):
• Reviews (n=6)
• PD studies with no 24 hour mean % of holding time at steady state (n=17)
• PD studies with no EE healing data (n=2)
• PK study with no pH holding time data (n=1)
• pH impedance study (n=1)
• Resistant EE (n=1)
• No EE healing data at 4 or 8 weeks (n=1)
• Protocol (n=1)
New studies included (n=30):
• pH holding time (n=18)
• EE healing rate (n=12)
Total studies included in review (n=181)
Reports of included studies (n=181):
• pH holding time (n=80)
• EE healing rate (n=101)

EE, erosive esophagitis; PD, pharmacodynamics; PK, pharmacokinetics.

- The 80 PD studies providing pH HTRs included 4,202 participants in 197 treatment arms.
- The 101 studies of erosive esophagitis included healing rate data for 42,155 subjects (intention-to-treat) in 192 treatment arms.
- Data for 13 individual drugs were included.

Relationship between pH >4 holding times and erosive esophagitis healing rates

- A non-linear mixed effects model meta-analysis was performed in NONMEM.
- The final model characterized and quantified the relationship between pH >4 holding times and erosive esophagitis healing rates using a maximum healing rate (E_{max}) model with logistic link function (**Figure 2**).
 - Two levels of nested random effects (on drug and treatment level) were characterized.
 - Time of the erosive esophagitis healing rate assessment (i.e., week 4 and/or 8) could only be implemented as a categorical covariate since those were the only timepoints available.
 - Additional significant covariates included the approval status (approved/not approved in the US and/or EU) on the healing rate sensitivity ($E_{HTR,50}$) and the between treatment variability, as well as drug class (H_2RA, PPI or P-CAB) on $E_{HTR,50}$.

Figure 2. Modeling the relationship between pH HTR and erosive esophagitis healing rates

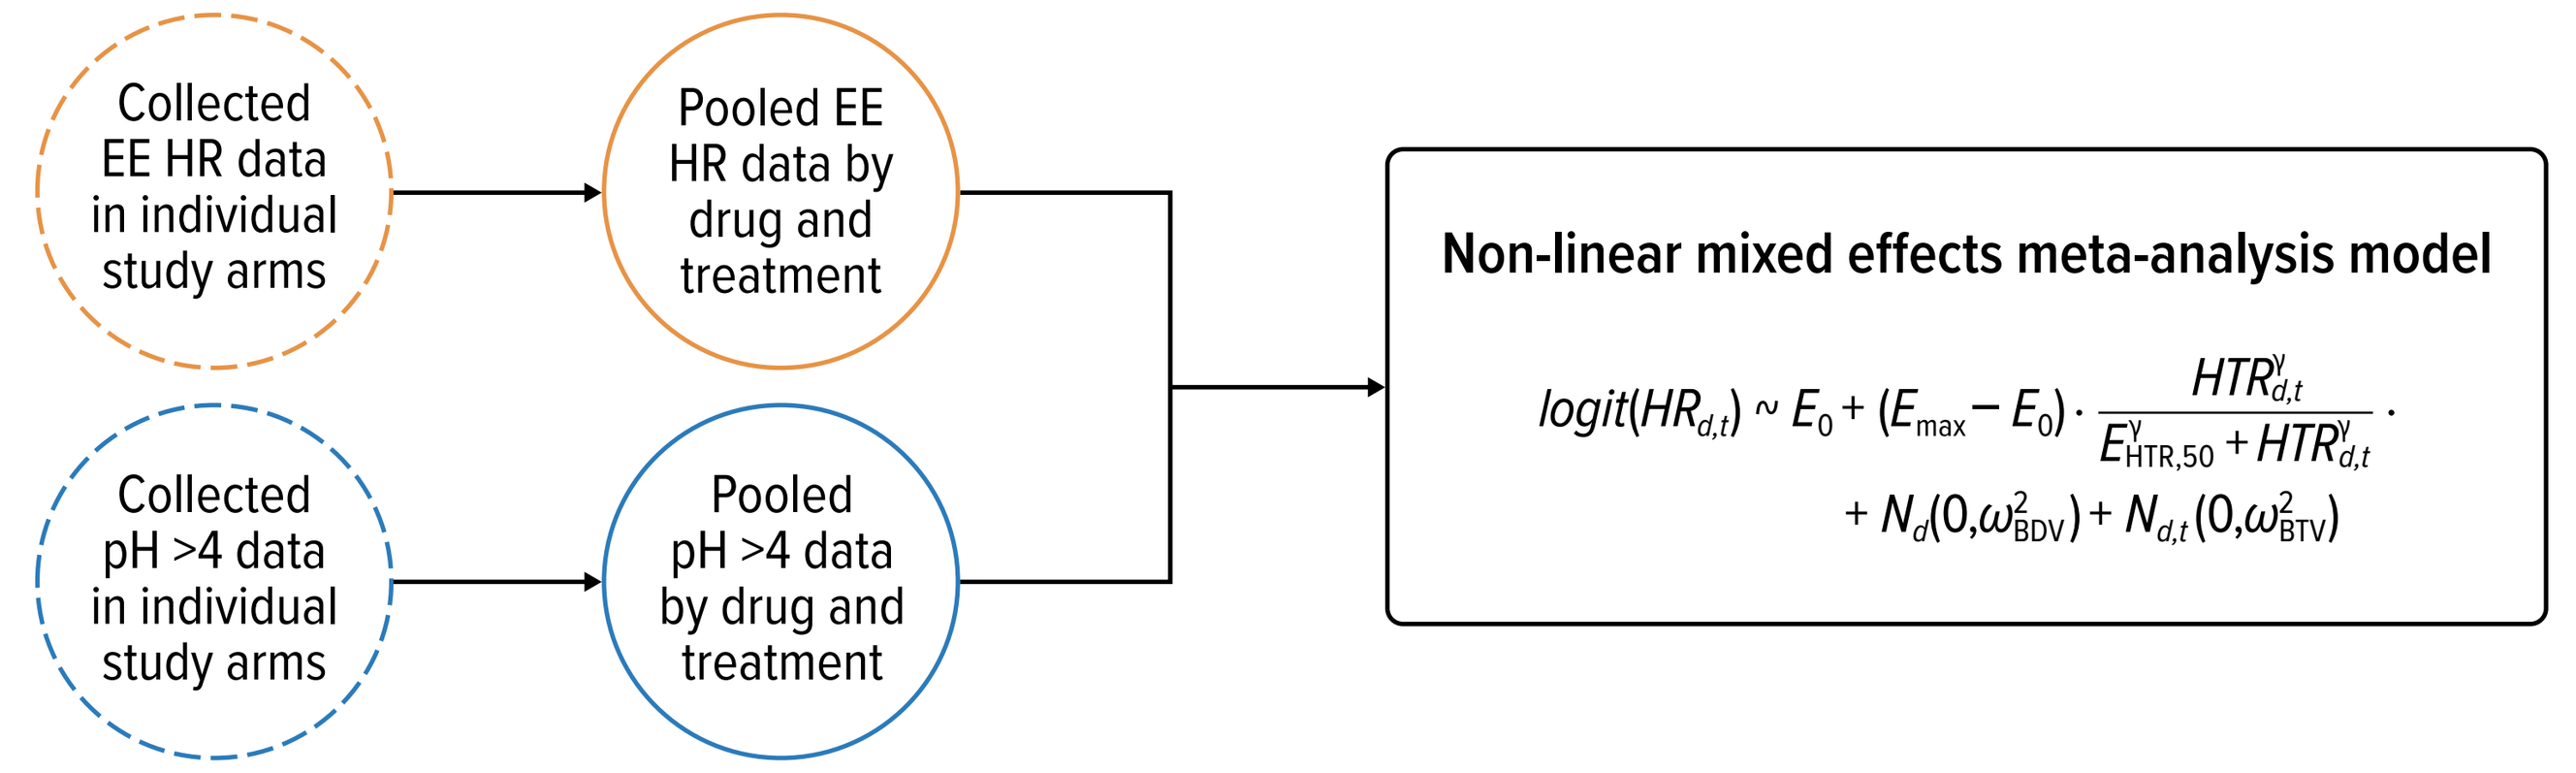
Collected EE HR data in individual study arms
Pooled EE HR data by drug and treatment
Collected pH >4 data in individual study arms
Pooled pH >4 data by drug and treatment
Non-linear mixed effects meta-analysis model
$logit(HR_{d,t}) \sim E_0 + (E_{max} - E_0) \cdot \frac{HTR_{d,t}^{\gamma}}{E_{HTR,50}^{\gamma} + HTR_{d,t}^{\gamma}} \cdot + N_d(0,\omega^2_{BDV}) + N_{d,t}(0,\omega^2_{BTV})$

BDV, between drug variability; BTV, between treatment variability; E_0, healing rate at 0% HTR; EE, erosive esophagitis; $E_{HTR,50}$, healing rate sensitivity; E_{max}: maximum healing rate; HR, healing rate; HTR, holding time ratio (mean time in hours/24 hours with pH >4).

- The meta-analysis model successfully characterized the relationship between pH >4 holding times and erosive esophagitis healing rates across all included drugs and doses.
- The final model adequately described the available data, and all model parameters could be estimated with good precision.
- Treatment duration and drug class significantly affected the healing rate sensitivity (**Figure 3**).
 - P-CABs were associated with higher efficacy range than H_2RAs and PPIs.
 - P-CABs achieved the longest periods of pH >4 for healing rates at all aggregated pH HTRs. P-CABs also achieved the highest erosive esophagitis healing rates at weeks 4 and 8.
 - P-CABs were predicted to attain median healing rates of approximately 90% by week 4, whereas healing rates for H_2RAs and PPIs increased through week 8 and remained lower than those of P-CABs.

Figure 3. Predicted erosive esophagitis healing rates

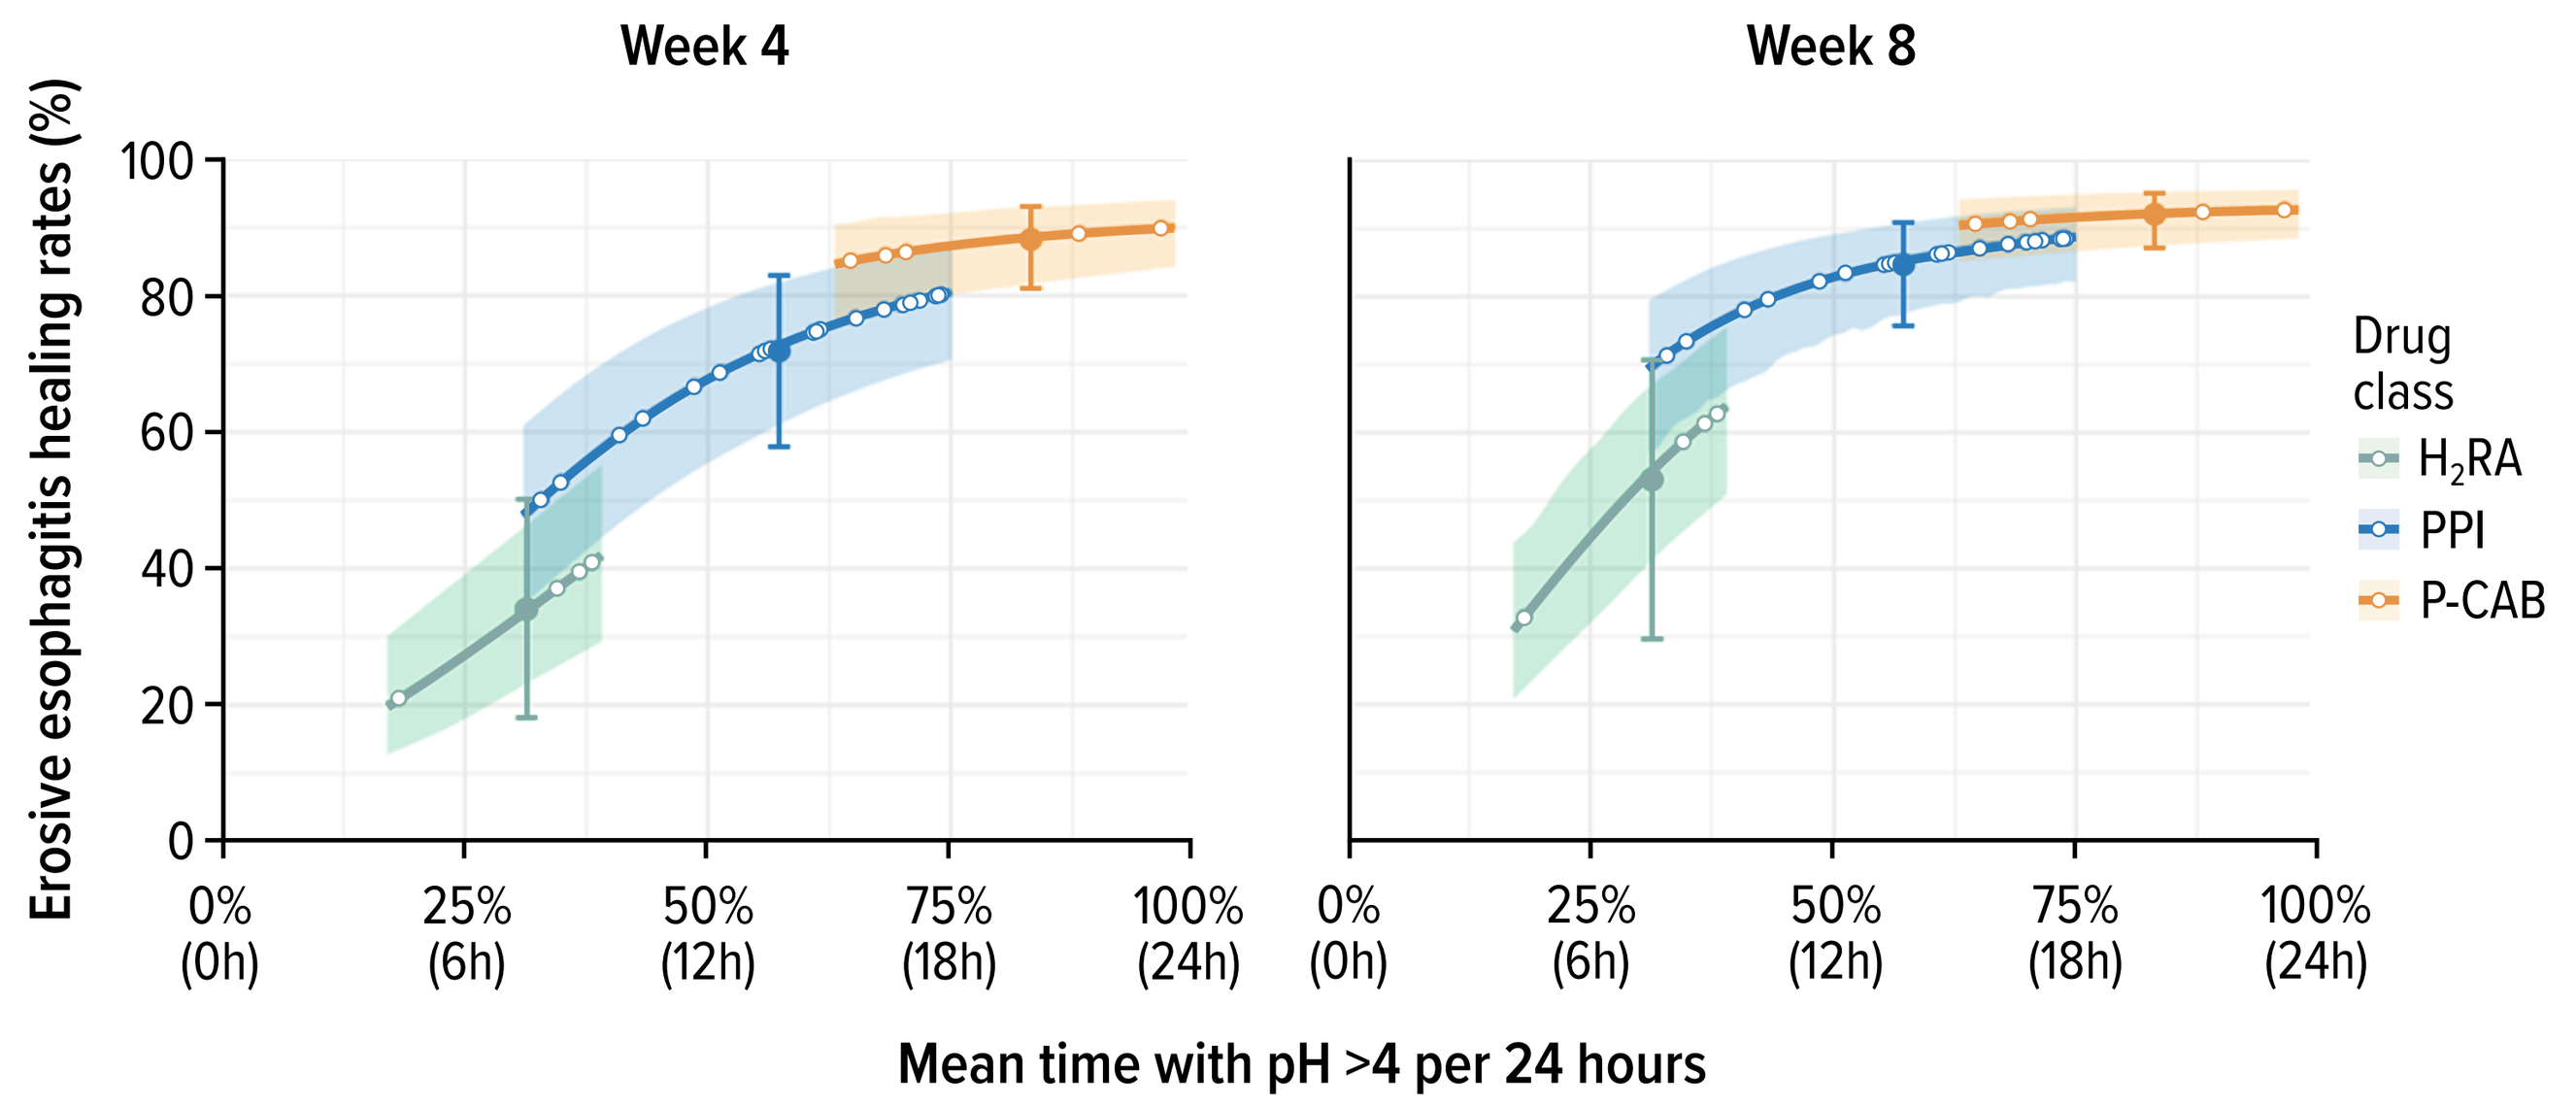
Week 4
Week 8
Erosive esophagitis healing rates (%)
0
20
40
60
80
100
0% (0h)
25% (6h)
50% (12h)
75% (18h)
100% (24h)
Mean time with pH >4 per 24 hours
Drug class
H_2RA
PPI
P-CAB

Predicted erosive esophagitis healing rates across drug classes in studies with approximately 100 patients included. Solid dots and error bars indicate medians and 80% prediction intervals averaged per drug class. Sigmoid line-segments and shaded areas indicate the typical model response and 80% prediction intervals from 2,000 simulations across pH HTRs. Open circles indicate individual predictions at aggregated pH HTR levels from the analysis data set.
h, hour; H_2RA, H_2-receptor antagonist; P-CAB, potassium-competitive acid blocker; PPI, proton pump inhibitor.

Probability of failing to achieve healing in erosive esophagitis

- We derived probabilities of failing to achieve healing rates of 80%, 85% and 90% with P-CABs versus H_2RAs or PPIs (**Figure 4; Table**) by means of clinical trial simulations.
- The predicted probabilities of failing to achieve 80–90% healing rates were lower for P-CABs than for H_2RAs or PPIs.
- The probability of failing to achieve 90% healing with P-CABs was 64.7% at week 4 and 27.5% at week 8; for H_2RAs it was 100% at weeks 4 and 8; for PPIs, it was 99.5% at week 4 and 84.3% at week 8.

Figure 4. Probabilities of failing to achieve target minimum erosive esophagitis healing rates of (A) 80%, (B) 85% or (C) 90% by drug class

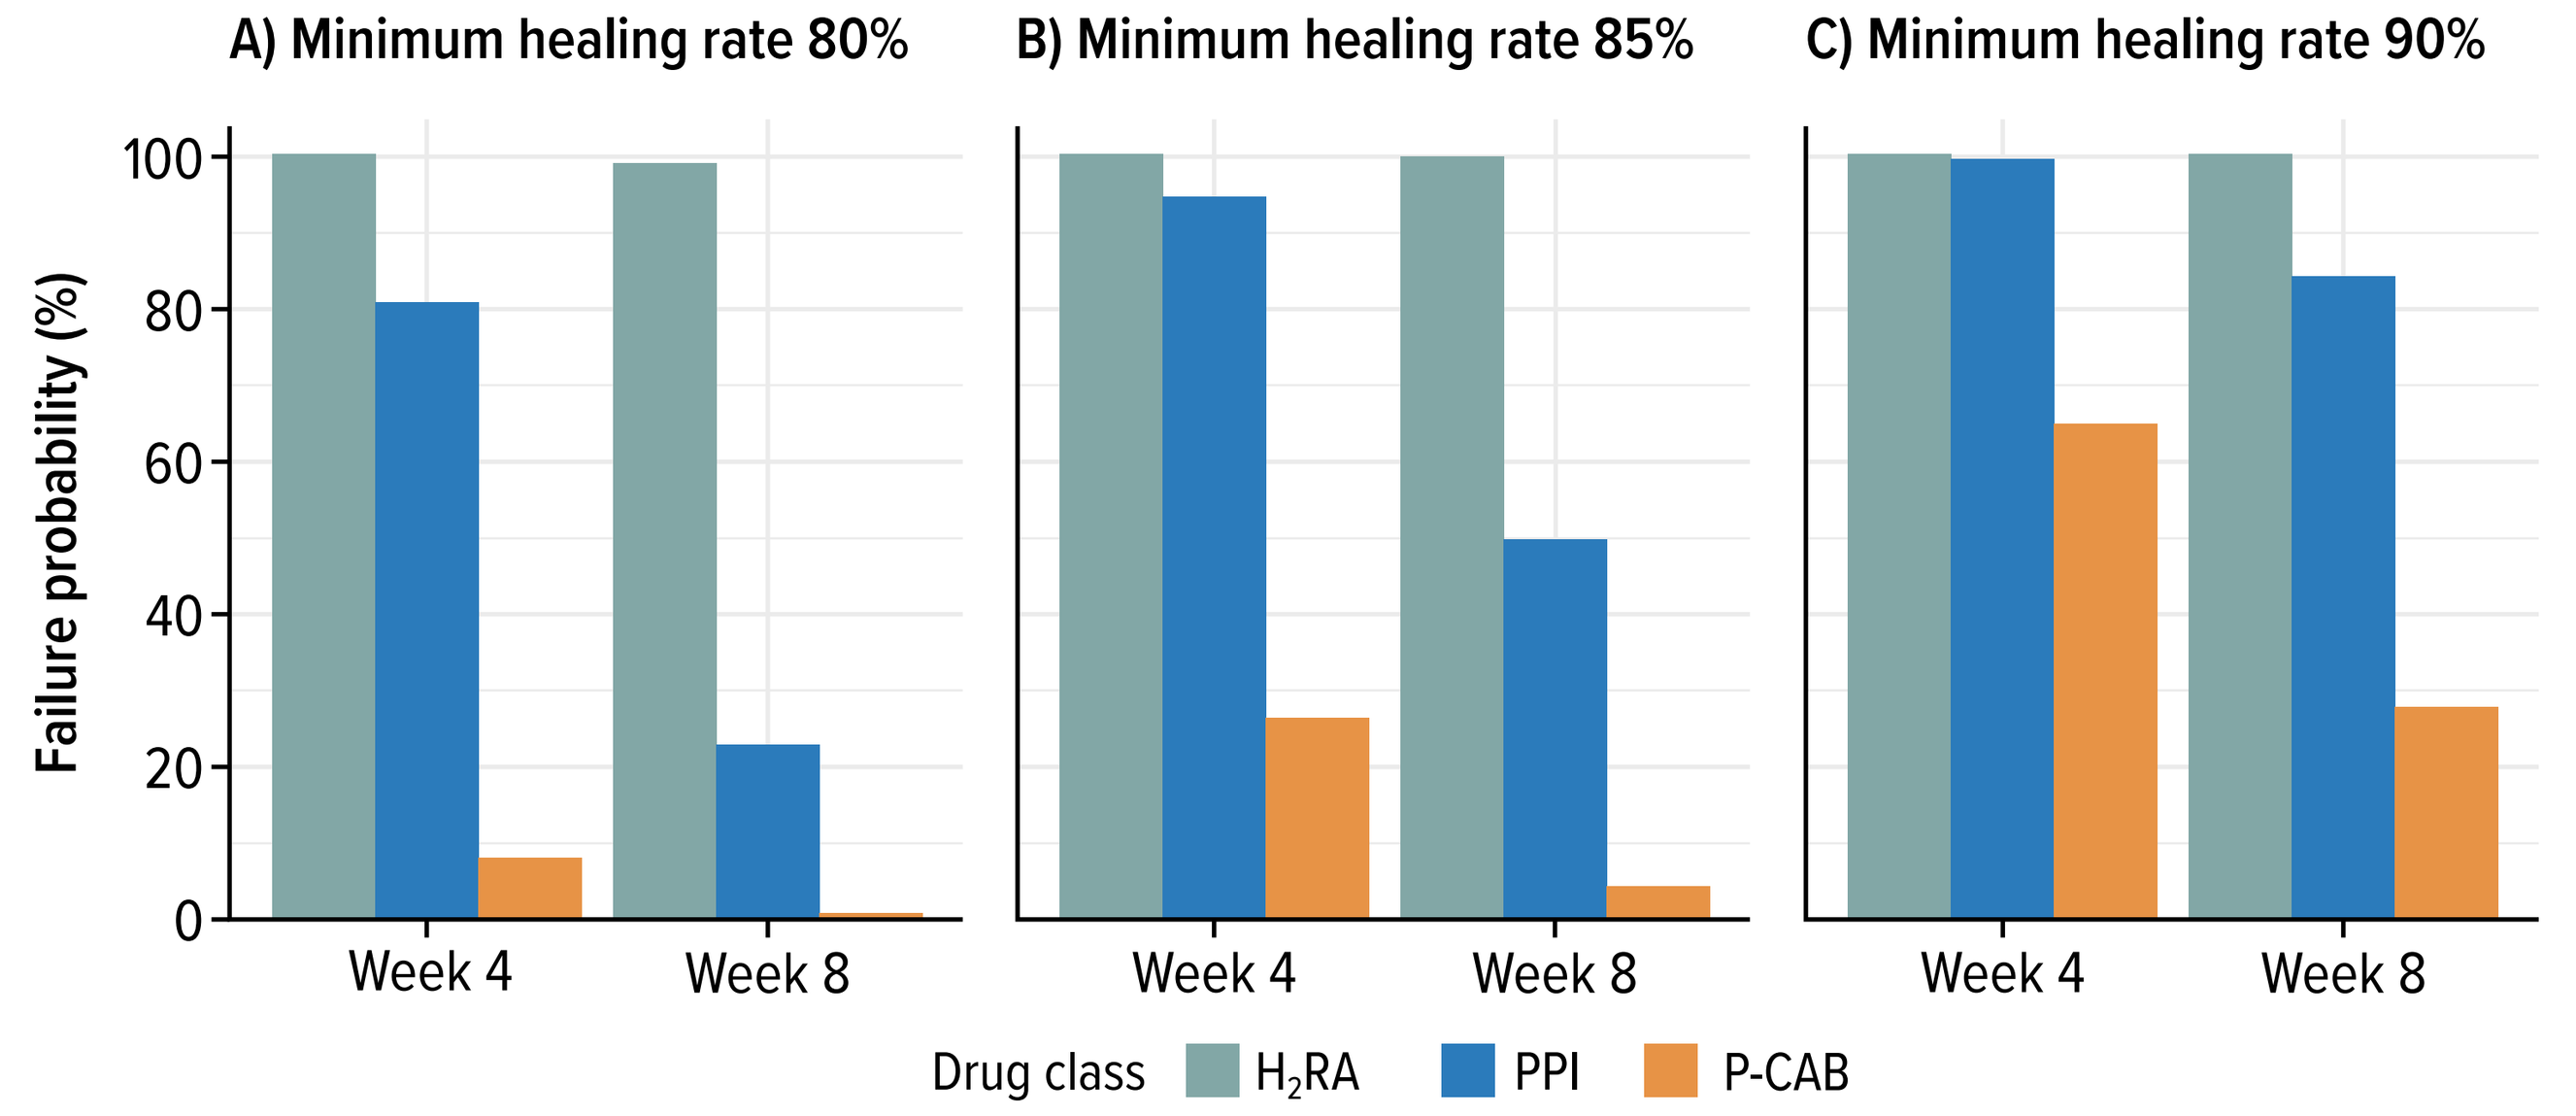
A) Minimum healing rate 80%
B) Minimum healing rate 85%
C) Minimum healing rate 90%
Failure probability (%)
0
20
40
60
80
100
Week 4
Week 8
Drug class
H_2RA
PPI
P-CAB

H_2RA, H_2-receptor antagonist; P-CAB, potassium-competitive acid blocker; PPI, proton pump inhibitor.

Table. Probabilities of failure to achieve target minimum erosive esophagitis healing rates

Target minimum erosive esophagitis healing rate	Drug class	Average time (h) observed with pH >4 per 24 h	Required time (h) with pH >4 to achieve target healing rate		Probability of failure (%)	
			Week 4	Week 8	Week 4	Week 8
80%	H_2RA	7.5	>24[a]	15.3	100.0	99.0
	PPI	13.7	17.7	10.5	80.7	22.6
	P-CAB	20.0	12.0	7.2	7.8	0.5
85%	H_2RA	7.5	>24[a]	19.5	100.0	99.9
	PPI	13.7	22.6	13.5	94.7	49.7
	P-CAB	20.0	15.4	9.2	26.2	4.2
90%	H_2RA	7.5	>24[a]	>24[a]	100.0	100.0
	PPI	13.7	>24[a]	20.5	99.5	84.3
	P-CAB	20.0	23.4	14.0	64.7	27.5

[a]Target healing rate cannot be reached.
h, hour; H_2RA, H_2-receptor antagonist; P-CAB, potassium-competitive acid blocker; PPI, proton pump inhibitor.

CONCLUSIONS

- **P-CABs provide a longer duration at pH >4, higher predicted erosive esophagitis healing rates, and lower probabilities of failure to achieve healing, than H_2RAs and PPIs.**
- **The more effective and prolonged acid suppression achieved by the P-CABs' mechanism of action appears to provide a meaningful clinical impact on healing of erosive esophagitis.**
- **Although this analysis is limited by the fact that data for HTR and erosive esophagitis were combined from different studies, the amount of pooled data increases confidence in the predictive value of the model.**

Acknowledgments

Medical writing support was provided by Samantha Agron, MD of Synergy Medical Communications, funded by Phathom Pharmaceuticals in accordance with Good Publications Practice (GPP 2022) guidelines.

Conflicts of interest

CWH has served as a consultant for Phathom Pharmaceuticals, RedHill Biopharma, Ironwood Pharmaceuticals and Allakos, and as a speaker for RedHill Biopharma, Alnylam Pharmaceuticals and Sanofi/Genzyme; he also owns stock in Antibe Therapeutics. **CS** has served as a consultant, an advisory board member, and a speaker for Phathom Pharmaceuticals, and as a speaker for Takeda. **EL** and **DJM** are employees of Phathom Pharmaceuticals; **EL** also discloses stockholder interest in Phathom Pharmaceuticals. **GL** and **AF** of thinkQ[2] were funded by Phathom Pharmaceuticals to undertake this work and have also served as consultants for Takeda. **RH** has served as a consultant for Takeda, Dr Reddy's Laboratories, Cinclus Pharma, Antibe Therapeutics and Phathom Pharmaceuticals, and owns stock in Antibe Therapeutics.

References

1. Scarpignato C et al. *BMC Med* 2016;14:179.
2. Hunt RH. *Arch Intern Med* 1999;159:649-57.
3. Mejia A and Kraft WK. *Expert Rev Clin Pharmacol* 2009;2:295-314.
4. Yuan Y and Hunt RH. *Gastroenterology* 2009:136 (Suppl).
5. Katz PO et al. *Aliment Pharmacol Ther* 2007;25:617-28.
6. Ashida K et al. *World J Gastroenterol* 2018;24:1550-61.
7. Fox RK and Muniraj T. *Med Clin North Am* 2016;100:827-50.
8. Shin JM and Sachs G. *Expert Rev Clin Pharmacol* 2009;2:461-8.
9. Laine L et al. *Am J Gastroenterol* 2022;117:1158-61.
10. Iwakiri K et al. *J Gastroenterol* 2022;57:267-85.
11. Zhang M, Xiao Y, Chen M. *Therap Adv Gastroenterol* 2022;15:17562848221122623.
12. Jenkins H et al. *Aliment Pharmacol Ther* 2015;41:636-48.
13. Sakurai Y et al. *Clin Transl Gastroenterol* 2015;6:e94.
14. Rawla P et al. *World J Gastrointest Pharmacol Ther* 2018;9:63-8.

Funding details

This study was sponsored by Phathom Pharmaceuticals.